MP0621 (CKIT X CD16A X CD47), A MULTI-SPECIFIC SWITCH-DARPIN WITH CONDITIONAL BLOCKADE OF CD47, TARGETING HEMATOPOIETIC STEM CELLS: PRECLINICAL EVALUATION OF A NEXT-GENERATION CONDITIONING AGENT FOR STEM CELL TRANSPLANTATION.

A. Link, T. Frasconi, S. Wullschleger, N. Venetz-Arenas, A. Ribeiro, A. Schlegel, Y. Kaufmann, A. Auge, W. Ali, N. Fic, Z. Siddiqui, A. Eggenschwiler, S. Jetzer, S. Häberle, D. Prekajski, H. De Winter, K. Dawson, A. Croset and A. Goubier

Molecular Partners AG, Zurich-Schlieren, Switzerland

INTRODUCTION

Current high-intensity myeloablative conditioning for hematopoietic stem cell transplantation (HSCT) is associated with toxicity which limits eligibility for this potentially curative treatment option for patients with acute myeloid leukemia (AML) and other malignant and non-malignant diseases. There is a high unmet need for less toxic conditioning regimens to allow broader access to HSCT for these patients.

- cKit (CD117), a critical receptor for hematopoietic stem cell maintenance, is an attractive target to selectively eliminate HSCs as well as leukemic stem cells
- Preclinical studies in mice suggest that cKit-targeting Abs alone do not sufficiently deplete HSCs for efficient HSCT and would benefit from concomitant blockade of the CD47 "do-not-eat-me" signal[1,2]
- In addition, activity of a-CD47 antibodies has been shown to depend on interaction with activating Fc-receptors[3]
- However, toxicities of a-CD47 Abs limit effective use of combining a cKit-targeting approach with systemic blockade of CD47

AIM

To achieve a safe and effective targeted conditioning by combining inhibition of cKit signaling with an active Fc-effector function and conditional blockade of CD47 on target cells in a single molecule to induce potent but selective depletion of cKit+ cells.

METHODS

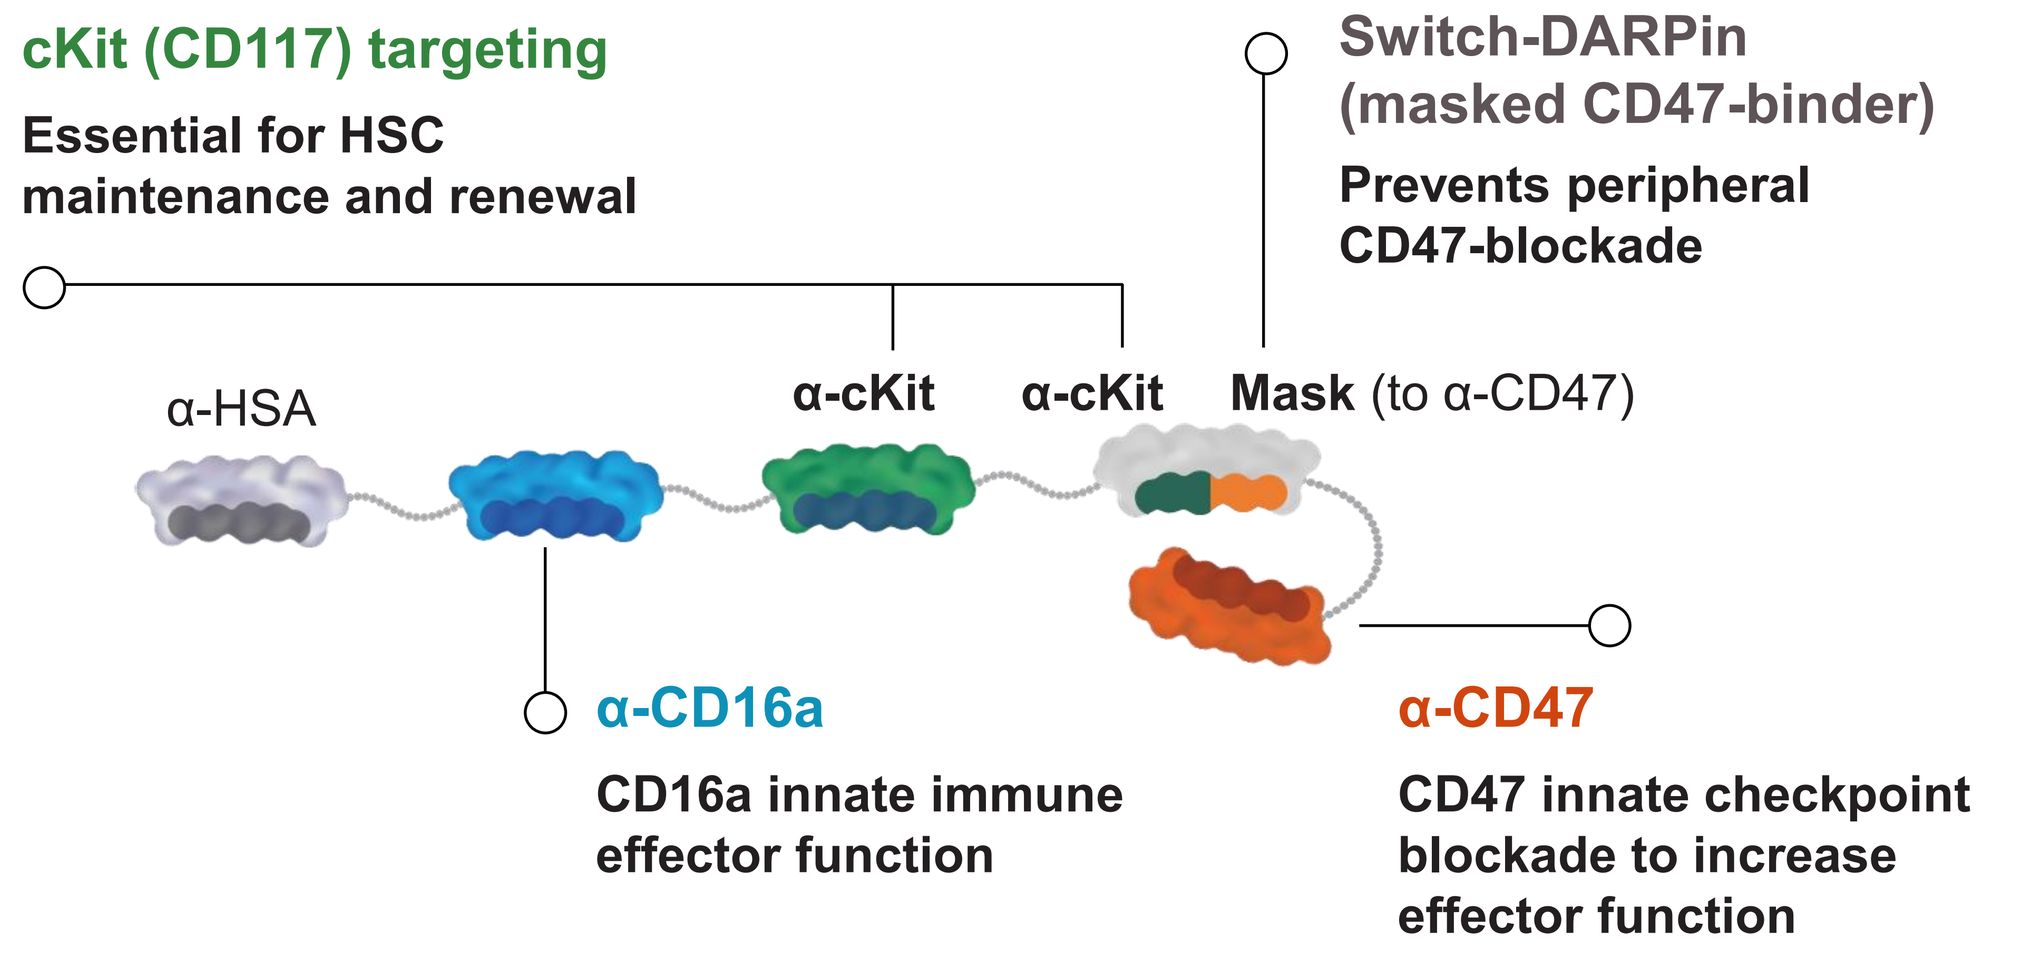
cKit (CD117) targeting
Essential for HSC maintenance and renewal
Switch-DARPin (masked CD47-binder)
Prevents peripheral CD47-blockade
α-HSA
α-cKit
α-cKit
Mask (to α-CD47)
α-CD16a
CD16a innate immune effector function
α-CD47
CD47 innate checkpoint blockade to increase effector function

- MP0621, a cKit x CD16a x CD47 Switch-DARPin was generated using our Designed Ankyrin-Repeat Protein platform.
- MP0621 contains a masked CD47 blocker that is released only upon binding to cKit and triggers conditional immune cell-mediated killing of HSCs by engaging macrophages and NK cells via CD16a (FcγR3a).
- In vitro activity was assessed by macrophage and NK cell assays.
- In vivo efficacy to deplete cKit⁺ cells in BM was assessed in a hCD34⁺ humanized NSG mouse model as well as in cynomolgus monkeys.

ACKNOWLEDGEMENTS

We would like to thank the entire team at Molecular Partners for their support and contribution.

CONTACT INFORMATION

info@molecularpartners.com / attention of A. Link

REFERENCES

1. Chhabra, A. et al. *Science translational medicine vol. 8,351 (2016):351ra105.*
2. Persaud, S.P. et al. *Blood advances vol. 8,17 (2024): 4502-4506.*
3. Osorio JC et al. Cancer Cell. 2023 Dec 11;41(12): 2051-2065
4. Kwon, H.S. et al. *Blood. 2019 May 9;133(19): 2104-2108.*

RESULTS

cKit-conditional blockade of CD47 induces enhanced phagocytosis of cKit⁺ target cells

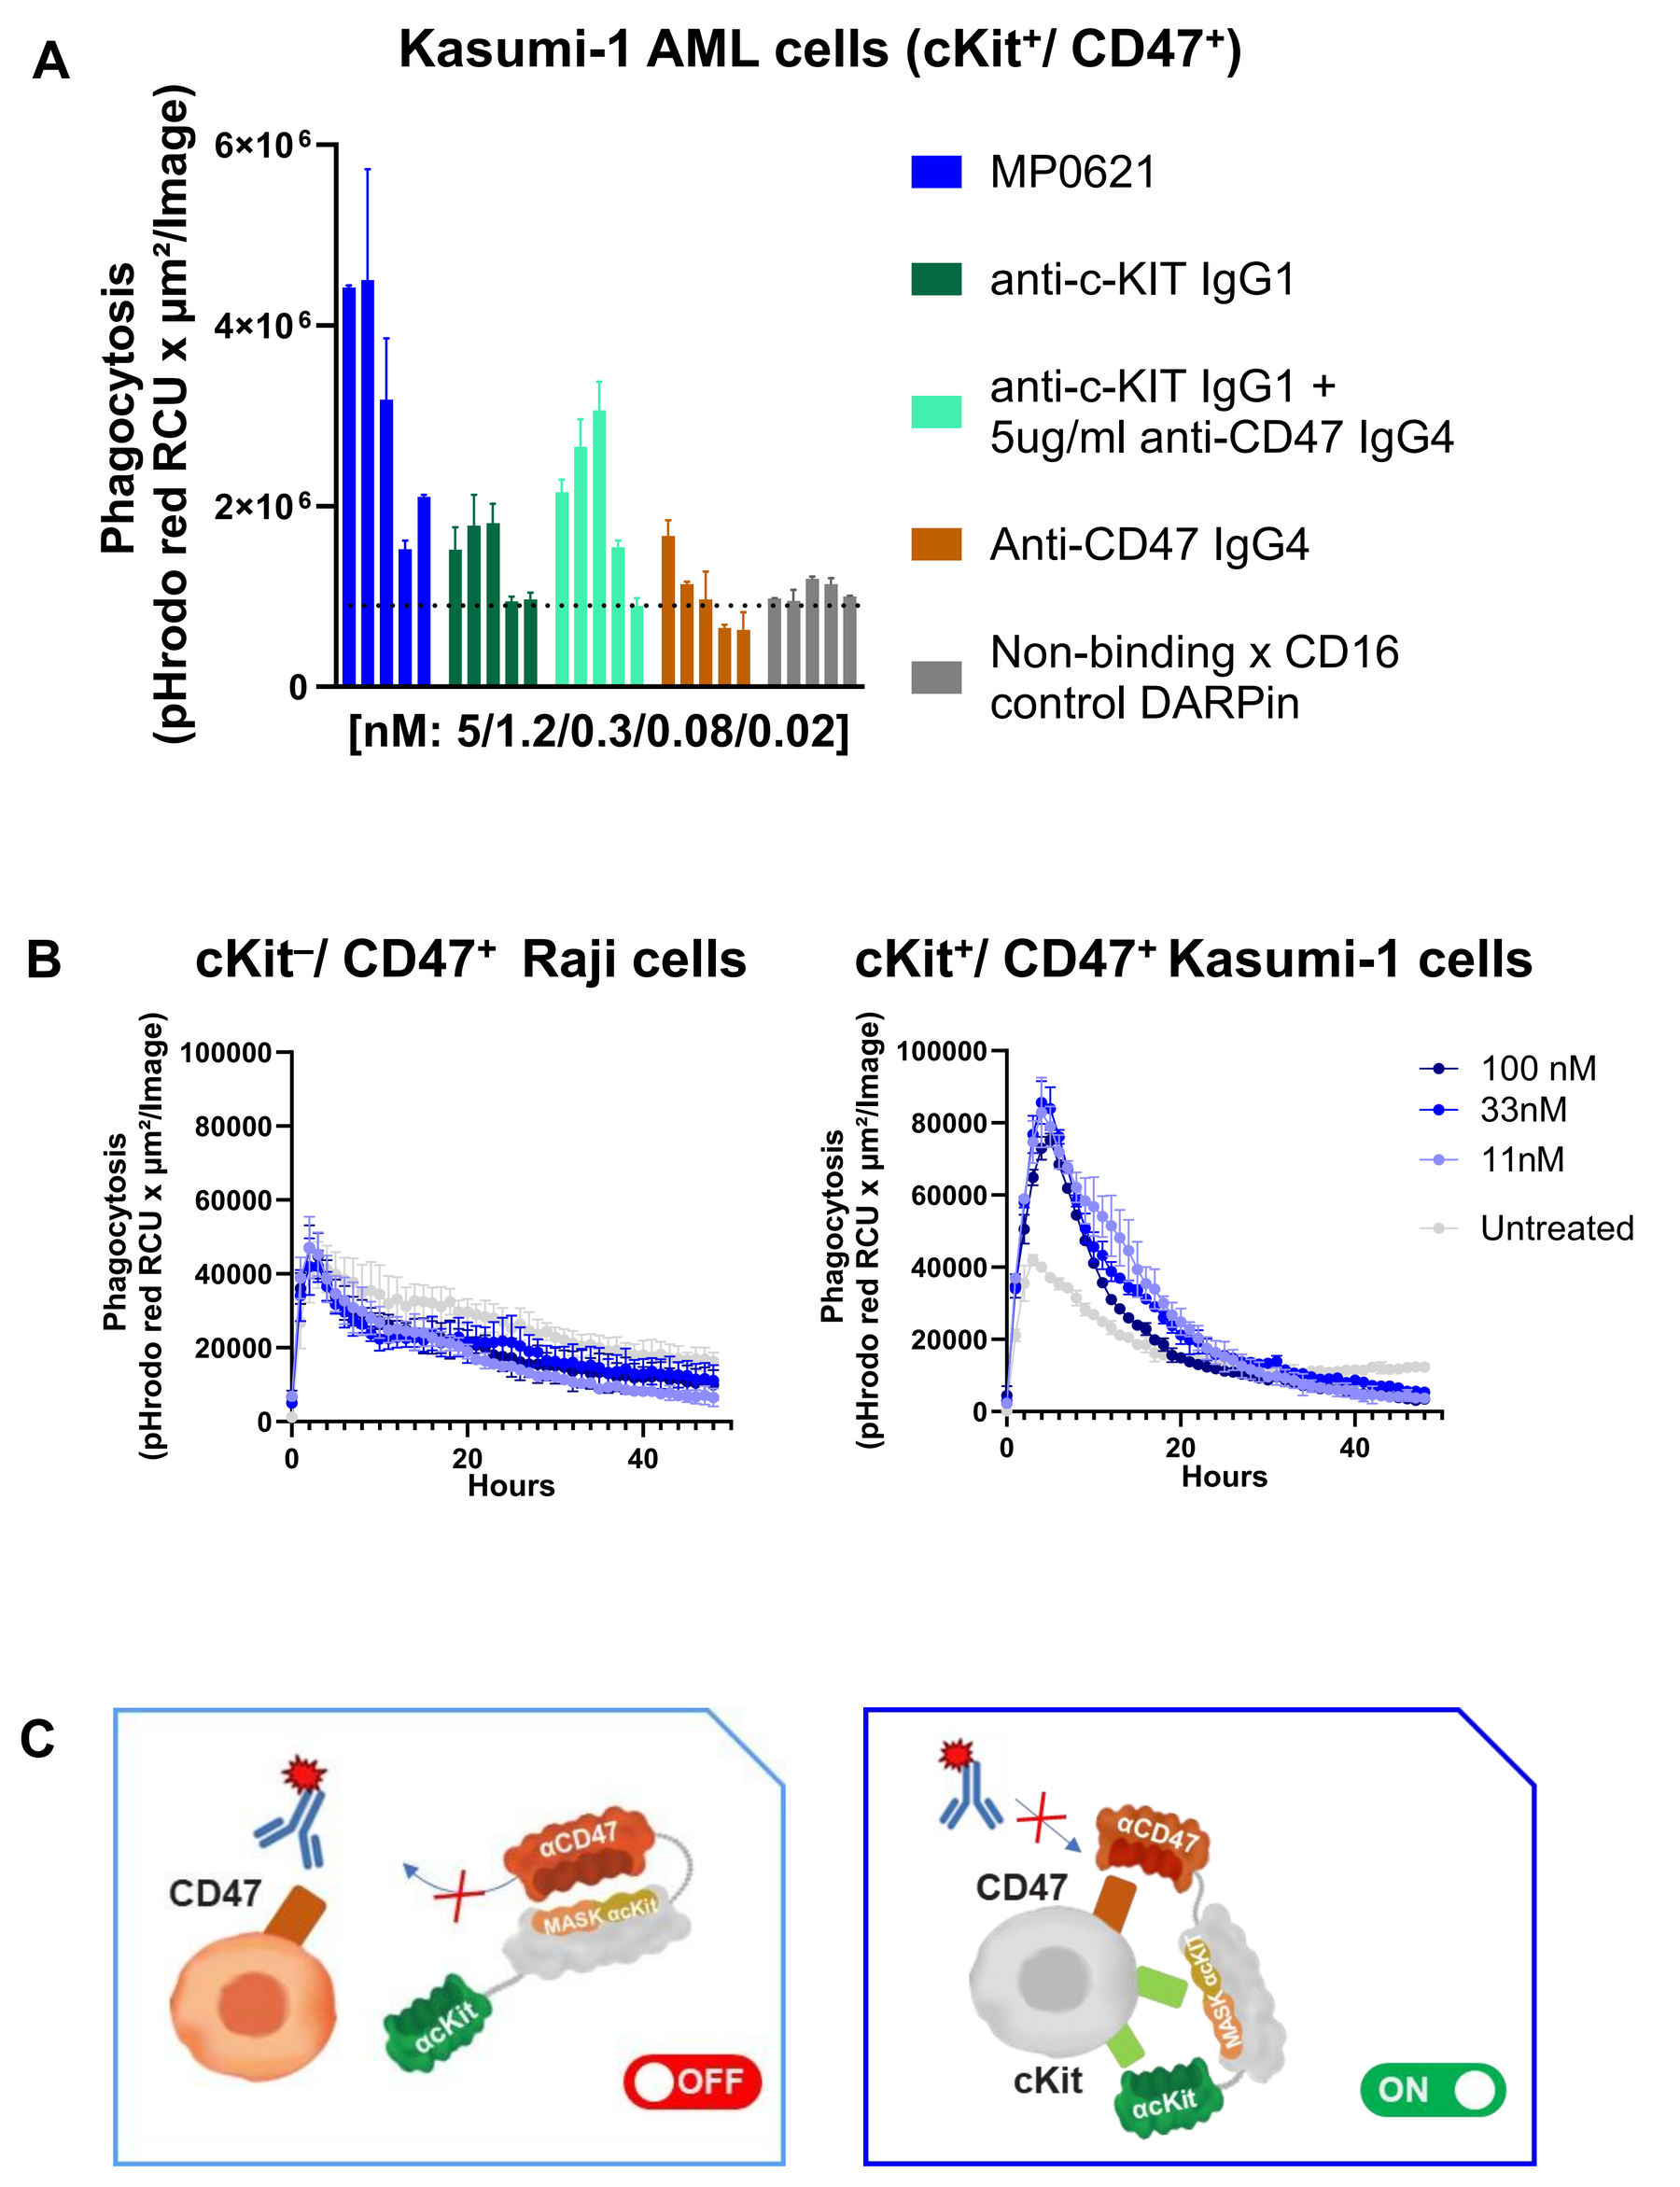
A
Kasumi-1 AML cells (cKit⁺/ CD47⁺)
Phagocytosis (pHrodo red RCU x µm²/Image)
[nM: 5/1.2/0.3/0.08/0.02]
MP0621
anti-c-KIT IgG1
anti-c-KIT IgG1 + 5ug/ml anti-CD47 IgG4
Anti-CD47 IgG4
Non-binding x CD16 control DARPin
B
cKit⁻/ CD47⁺ Raji cells
cKit⁺/ CD47⁺ Kasumi-1 cells
Hours
100 nM
33nM
11nM
Untreated
C
CD47
cKit
αCD47
αcKit
OFF
ON

Figure 1. MP0621 induces macrophage-mediated phagocytosis comparable/ superior to a combination of a-cKit and a-CD47 Abs of cKit⁺ target cells, while preventing phagocytosis of cKit⁻ CD47⁺ cells

*(**A**) Switch-DARPin induced phagocytosis of pHrodo-labelled cKit-expressing AML cell line Kasumi-1 mediated by human monocyte-derived M0 MF at an E:T ratio of 1:4. The plots show the AUC obtained upon 48-h co-culture in presence of increasing concentrations of MP0621, α-cKit Ab (Fc-active version (IgG1) of JSP-191, reproduced in house) or a combination of a-cKit and a-CD47 Abs. Representative example of at least three independent experiments.*

*(**B**) MP0621-induced phagocytosis of pHrodo-labelled cKit⁻ Raji cell line and cKit⁺ AML cell line Kasumi-1 at ascending concentrations of MP0621 mediated by human monocyte-derived M0 MF at an E:T ratio of 1:4. The plots show the signal obtained upon 48 h co-culture with the indicated amounts of DARPins. Representative example of at least three independent experiments.*

*(**C**) Schematic illustration of cKit-conditional CD47 engagement by MP0621 (CD47 switch 'off' in absence and 'On' in presence of cKit) reflecting the target cells shown in (B).*

MP0621 is able to induce cytotoxicity of human donor mobilized CD34⁺ HSCs

Figure 2. MP0621 induces NK cell mediated cytotoxicity of CD34⁺ HSCs from human donors. *NK cell mediated cytotoxicity assay of CD34⁺ mobilized cells from healthy human donors. The plot shows specific lysis obtained upon 24 h co-culture of PBMC-purified NK cells and CD34⁺ CD47⁺ cKit⁺ mobilized cells from healthy donors at an E:T ratio 5:1 in the presence of MP0621 and control DARPins. Representative example of five independent experiments.*

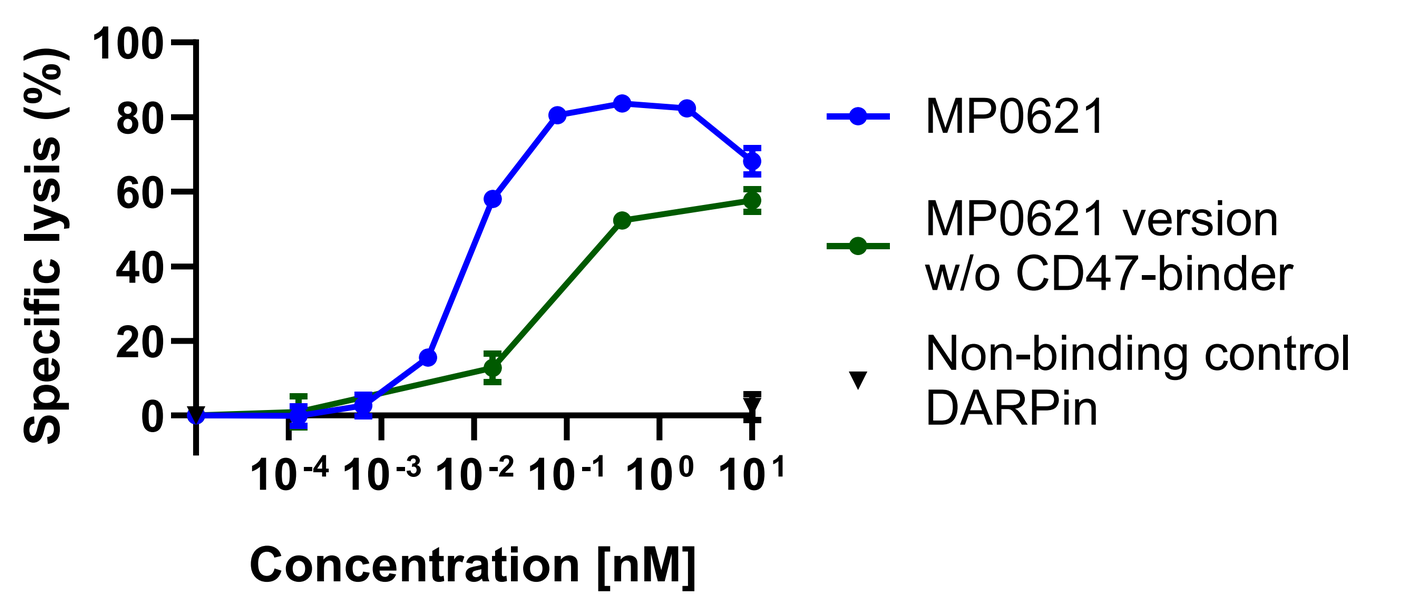
Specific lysis (%)
Concentration [nM]
MP0621
MP0621 version w/o CD47-binder
Non-binding control DARPin

MP0621 targets cKit⁺ cells in BM without depleting blood hCD45⁺ cells in hCD34⁺ NSG mice

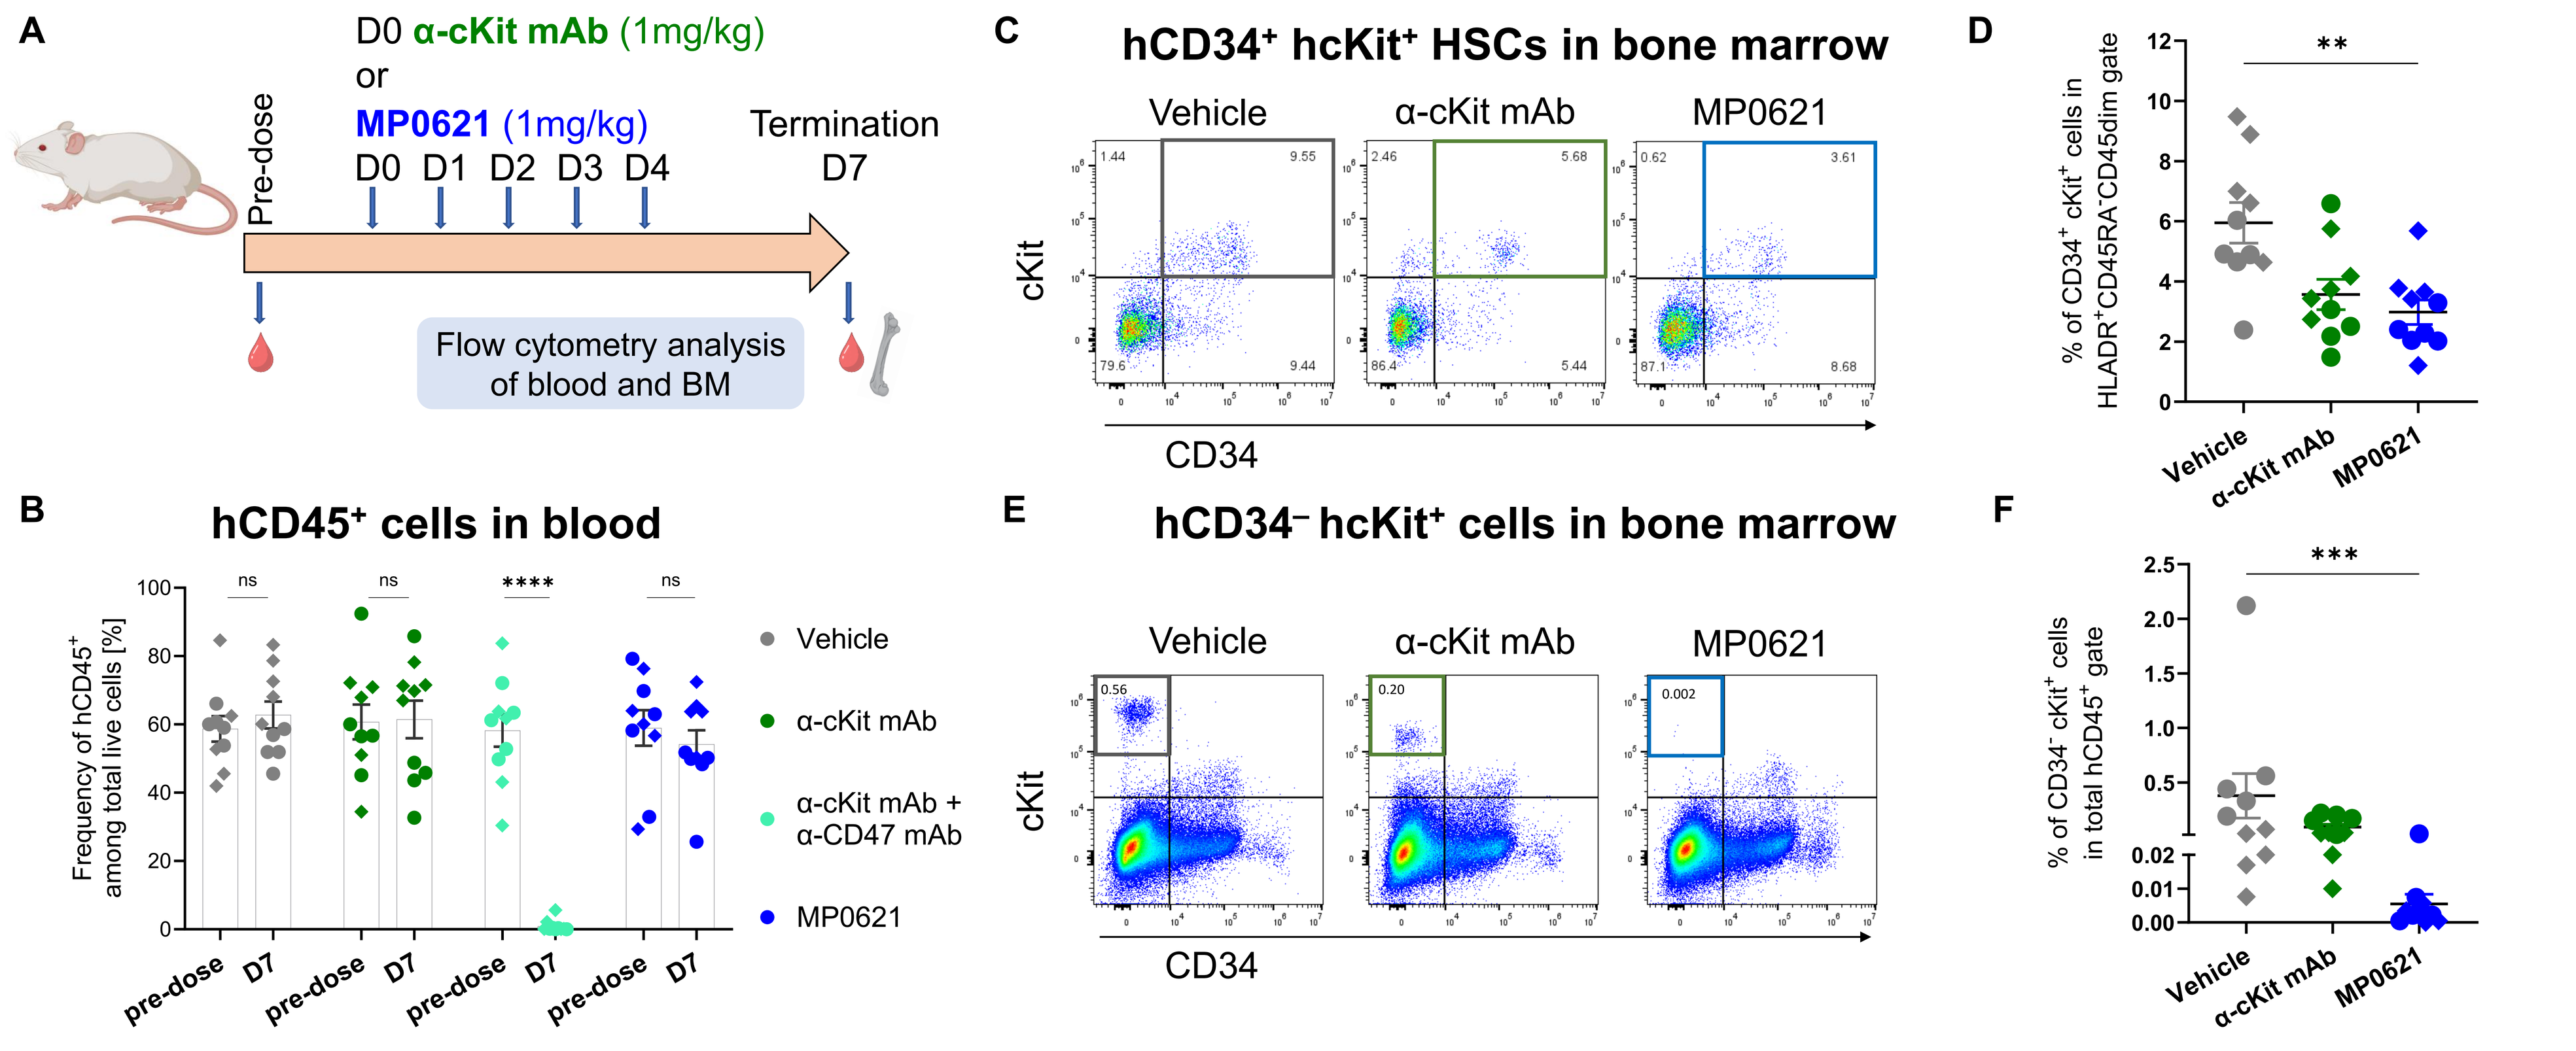
A
D0 α-cKit mAb (1mg/kg)
or
MP0621 (1mg/kg)
D0 D1 D2 D3 D4
Termination
D7
Pre-dose
Flow cytometry analysis of blood and BM
B
hCD45⁺ cells in blood
Frequency of hCD45⁺ among total live cells [%]
pre-dose
D7
Vehicle
α-cKit mAb
α-cKit mAb + α-CD47 mAb
MP0621
C
hCD34⁺ hcKit⁺ HSCs in bone marrow
Vehicle
α-cKit mAb
MP0621
cKit
CD34
E
hCD34⁻ hcKit⁺ cells in bone marrow
D
% of CD34⁺ cKit⁺ cells in HLADR⁺CD45RA⁻CD45dim gate
F
% of CD34⁻ cKit⁺ cells in total hCD45⁺ gate

Figure 3. Treatment with MP0621 targets human cKit⁺ HSCs engrafted in CD34⁺ humanized NSG mice, while preventing elimination of peripheral hCD45⁺ cells observed with unconditional a-CD47 blockade

*(**A**) Study design*

*(**B**) Flow cytometry analysis of hCD45⁺ cells in peripheral blood before and after treatment with MP0621, a-cKit mAb (JSP-191, N297Q-aglycosyl-Fc reproduced in house), or a combination of α-cKit mAb and α-CD47 (IgG4) mAbs. Displayed data are calculated as in the following equation: %hCD45⁺ = (100*%hCD45⁺)/ (%mCD45⁺ +%hCD45⁺); (Mann-Whitney test ****p<0.0001, ***p=0.007).*

*(**C-F**) Flow cytometry assessment of human cKit+ cell populations in the bone marrow of hCD34-humanized NSG mice treated with MP0621 or α-cKit mAb vs vehicle (**C**) Representative flow cytometry dot plots and (**D**) frequency of human hCD34⁺ hcKit⁺ with HSC signature (hCD45dim CD45RA⁻ HLADR⁺) (Kruskal-Wallis test ***p=0.004). (**E**) Representative flow cytometry plots and (**F**) frequency of BM hCD34⁻ hcKit⁺ cell population of total hCD45⁺ cells. (Kruskal-Wallis test **p=0.002; ***p=0.0004). Round and square symbols depict mice humanized with hCD34⁺HSCs isolated from cord blood of two different donors.*

MP0621 is cyno cross-reactive but effect on conditional CD47 blockade is reduced

A

		KD [M]	
Molecule	Target	human	cyno
MP0621	c-Kit	2.16E-11	2.59E-11
MP0621	CD16	1.50E-08	2.09E-08
α-CD47 domain	CD47	4.31E-10	3.29E-10

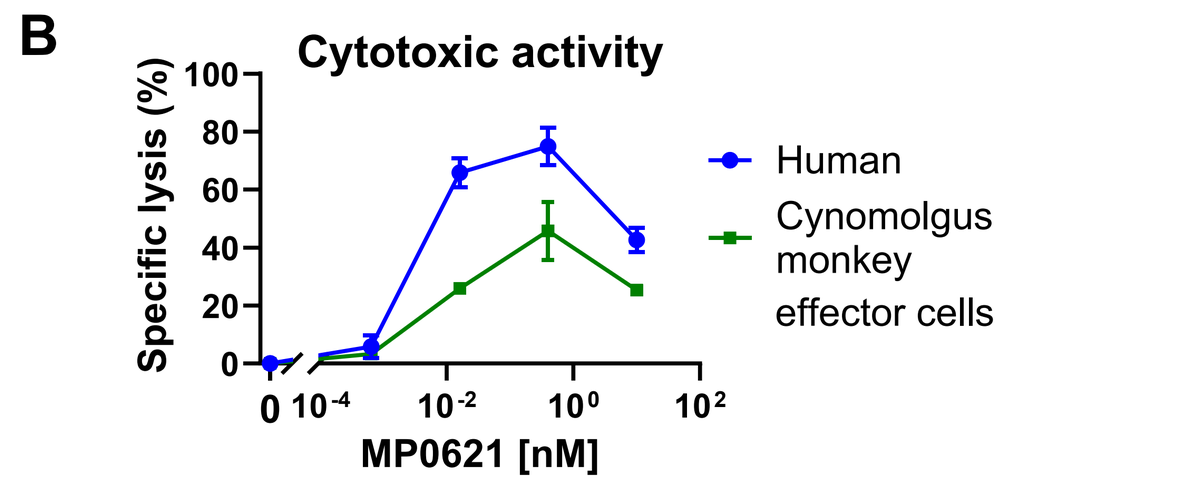
B
Cytotoxic activity
Specific lysis (%)
MP0621 [nM]
Human
Cynomolgus monkey effector cells

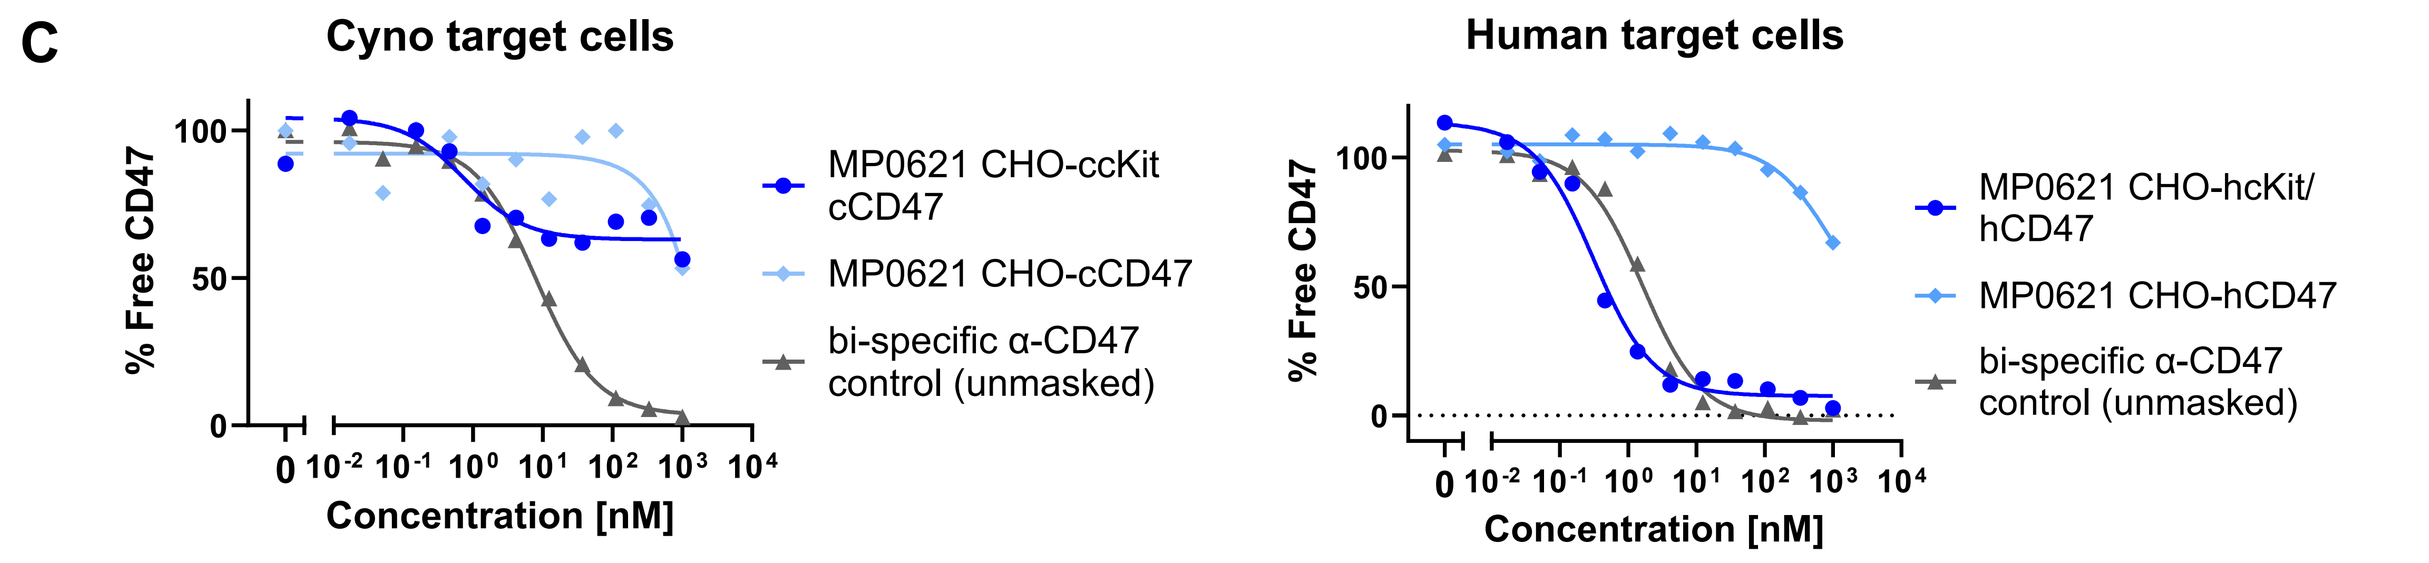
C
Cyno target cells
% Free CD47
Concentration [nM]
MP0621 CHO-ccKit cCD47
MP0621 CHO-cCD47
bi-specific α-CD47 control (unmasked)
Human target cells
MP0621 CHO-hcKit/ hCD47
MP0621 CHO-hCD47
bi-specific α-CD47 control (unmasked)

Figure 4. Cross-reactivity and in vitro bioactivity to human and cynomolgus monkey targets. *(**A**) KD values measured by surface-plasmon resonance. (**B**) In vitro killing of CD34⁺ mobilized cells from human donors by cyno CD16⁺ or human CD56⁺CD16⁺ effector cells. Plot shows target cell lysis upon 24 h coculture at an E:T ratio 5:1 in the presence of MP0621. Representative example of two independent experiments. (**C**) cKit-dependent conditional blockade of CD47 was measured using target cells engineered to express either cynomolgus monkey cKit and/or CD47 (CHO-cKit/ cCD47 or CHO-cCD47) or CHO cells expressing the human targets (CHO-hcKit/hCD47 or CHO-hCD47). Following incubation with MP0621 and washing, a biotinylated α-CD47 detection agent was added to the cells, followed by streptavidin-AF647. The signal obtained reflects the level of free CD47 available on the cell surface.*

MP0621 has a good safety profile in cynomolgus monkeys and transiently removes cKit^high BM cells

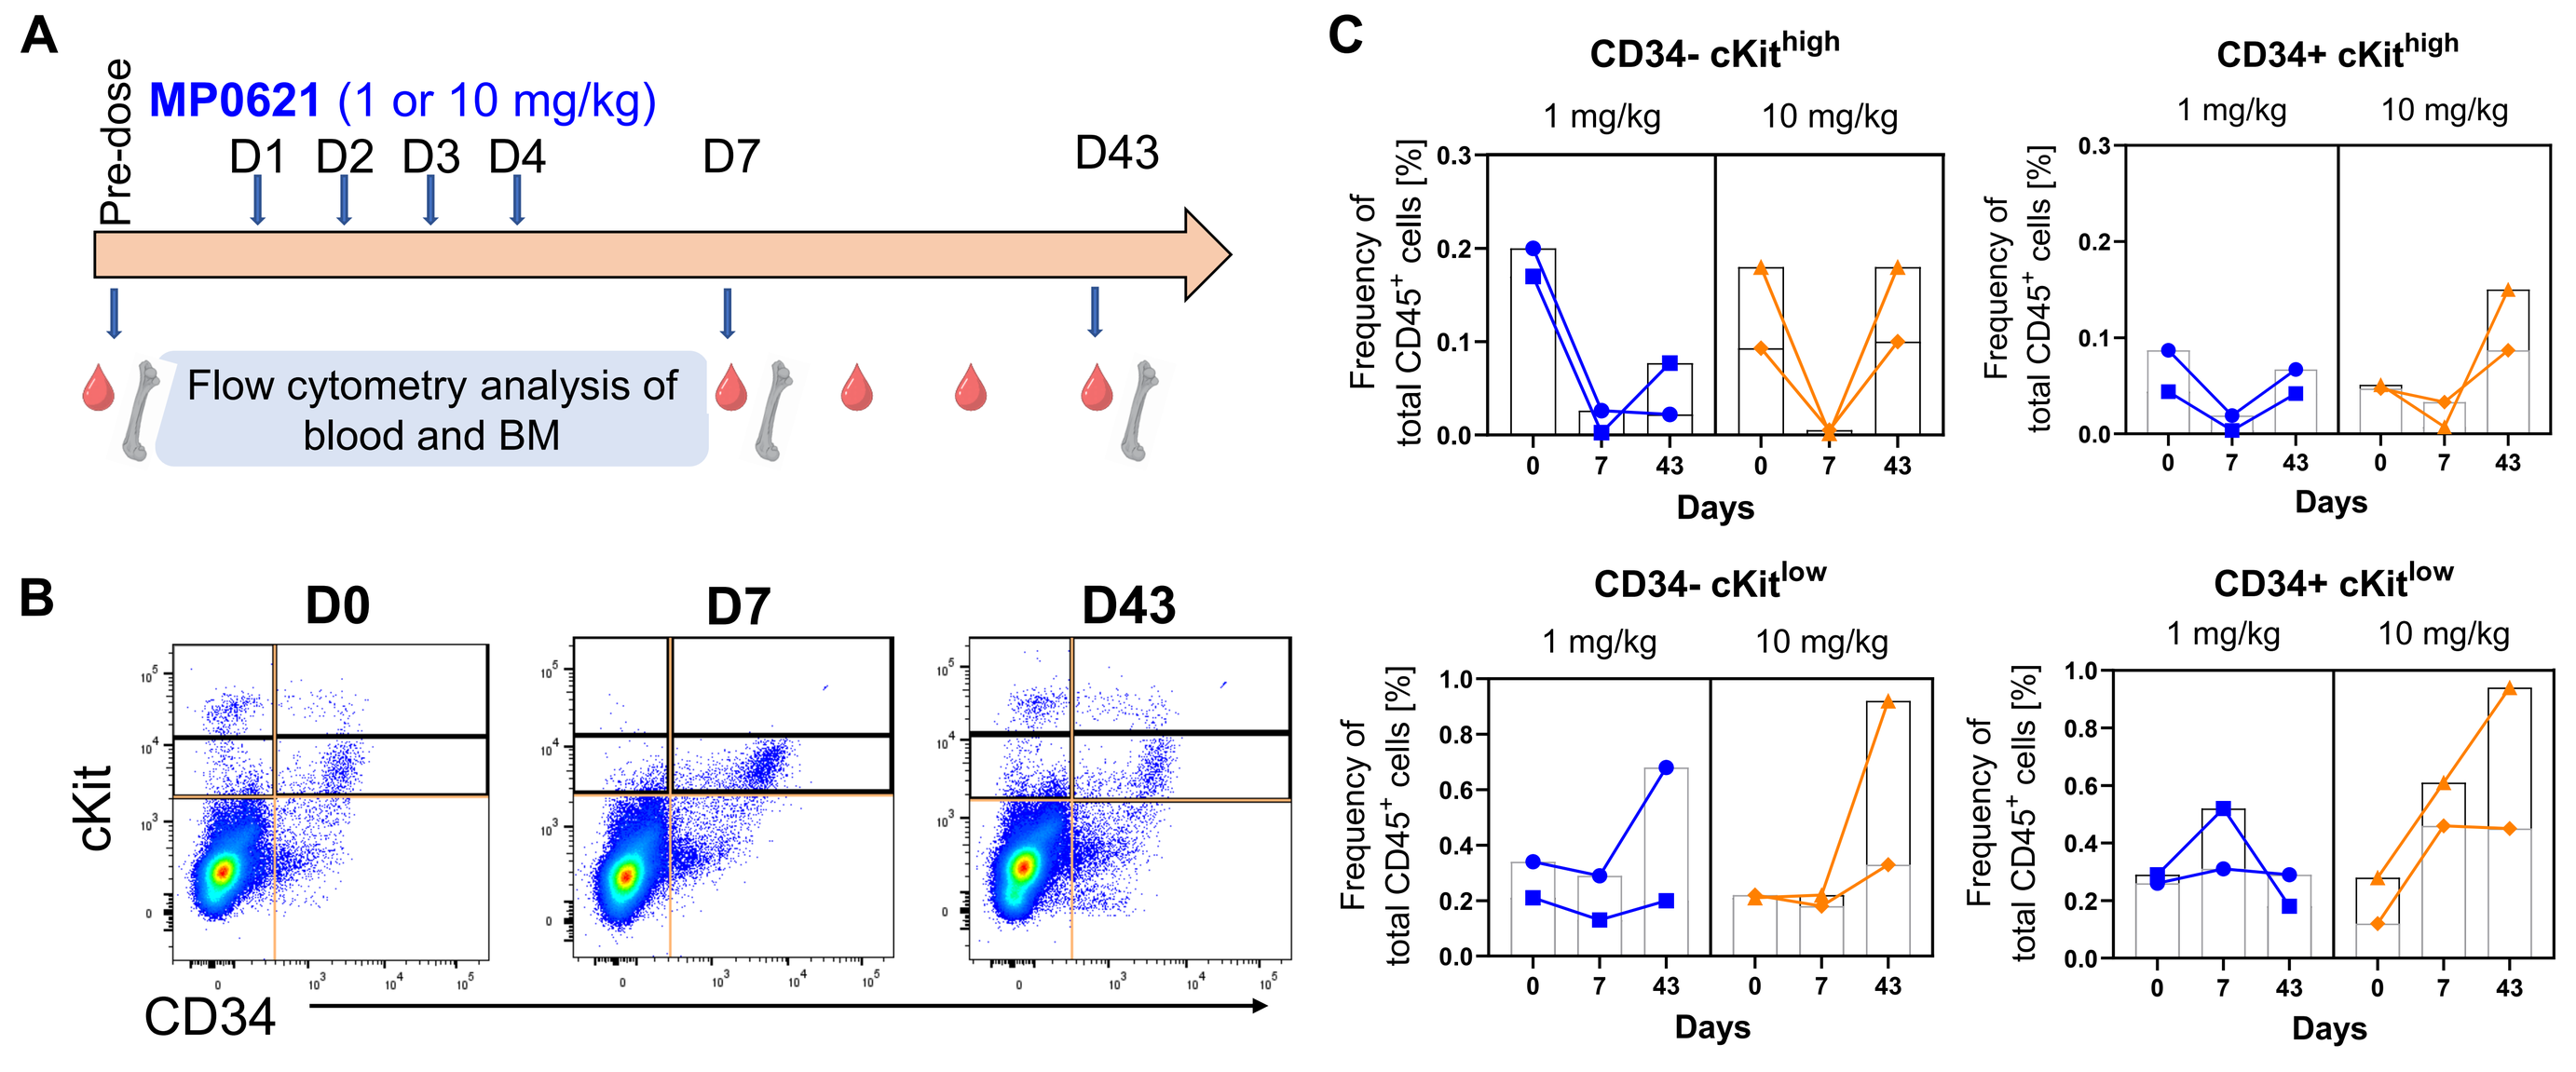
A
MP0621 (1 or 10 mg/kg)
Pre-dose
D1 D2 D3 D4
D7
D43
Flow cytometry analysis of blood and BM
B
D0
D7
D43
cKit
CD34
C
CD34- cKithigh
CD34+ cKithigh
CD34- cKitlow
CD34+ cKitlow
1 mg/kg
10 mg/kg
Frequency of total CD45⁺ cells [%]
Days

Figure 5. Treatment with MP0621 targets cKit⁺ cells in the bone marrow of cynomolgus monkeys. *Cynomolgus monkeys were treated for 4 days with 2 doses of MP0621 (1 and 10 mg/kg; 2 animals each). Bone marrow samples were analyzed for HSC depletion pre-dose (day 0) and on days 7 and 43. Blood parameters were assessed 3 times per week. (**A**) Study design (**B, C**) Flow cytometry analysis of cKit⁺ cell populations in the bone marrow of cynomolgus monkeys (2 animals per dose level) before and after treatment with 4 daily doses of 1 or 10 mg/kg of MP0621. (**B**) Representative flow cytometry dot plots (one animal from the 1 mg/kg dose group) and (**C**) frequency of cKit-expressing CD34⁺ and CD34⁻ cells of total CD45⁺ cells in the bone marrow.*

CONCLUSIONS

- **In summary, our preclinical results indicate that combining cKit-targeting with conditional blockade of CD47 in a single molecule using our Switch-DARPin platform might render the combination treatment a viable approach for HSC depletion in patients.**
- **The blockade of CD47 exclusively on target cells allows MP0621 to enhance efficacy of cKit-targeting, while reducing off-target effects seen with systemic anti-CD47 blockade.**
- **Thus, MP0621 represents a potential novel conditioning regimen that could improve the benefit risk profile of current HSCT conditioning strategies for patients.**

ABBREVIATIONS

mAb, (monoclonal) antibody; AML, acute myeloid leukemia; AUC, area under the curve; BM, bone marrow; D, day; ELISA, enzyme-linked immuno-sorbent assay; E:T, effector to target; FACS, fluorescence-activated cell sorting; MAC, myeloablative conditioning; HSC(T), hematopoietic stem cell (transplantation); HSA, human serum albumin; i.v., intravenous; LLOQ, lower limit of quantification; LSC, leukemic stem cell; MF, macrophage.